Outcomes and nursing management of advanced prostate cancer in older men treated with enzalutamide

Monique Williams,[1] Tamara Huebner,[2] Jeffrey Holzbeierlein,[3] Julie N. Graff,[4] Ugo De Giorgi,[5] Russell Szmulewitz,[6] Andrew J. Armstrong,[7] Arnulf Stenzl,[8] Cora N. Sternberg,[9] Jennifer Sugg,[10] Betty Thompson,[11] Neal D. Shore[12]

[1]Leidos Biomedical Research, Inc., Bethesda, MD, USA; [2]University of Michigan Rogel Cancer Center, Ann Arbor, MI, USA; [3]The University of Kansas Medical Center, Kansas City, KS, USA; [4]VA Portland Health Care System, Portland and Knight Cancer Institute, Oregon Health & Science University, Portland, OR, USA; [5]IRCCS Istituto Romagnolo per lo Studio dei Tumori (IRST) Dino Amadori, Meldola, Italy; [6]The University of Chicago, Chicago, IL, USA; [7]Duke Cancer Institute, Center for Prostate & Urologic Cancers, Durham, NC, USA; [8]University Hospital, Eberhard Karls University of Tübingen, Tübingen, Germany; [9]Englander Institute for Precision Medicine, Weill Cornell Medicine, Meyer Cancer Center, New York, NY, USA; [10]Astellas Pharma US, Northbrook, IL, USA; [11]Pfizer, San Francisco, CA, USA; [12]Carolina Urologic Research Center, Myrtle Beach, SC, USA

Objective

To review the outcomes and safety of enzalutamide plus androgen deprivation therapy (ADT) in older adults (≥75 years of age) with metastatic hormone-sensitive prostate cancer (mHSPC), and nursing implications for this population

Context

In a *post hoc* analysis of the ARCHES trial (NCT02677896), enzalutamide plus ADT was found to provide clinical benefit and to be generally well-tolerated in patients with mHSPC ≥75 years of age

Key Nursing Implications

Nurses can provide care that allows patients ≥75 years of age to benefit from mHSPC treatment options (such as enzalutamide) associated with improved survival and other outcomes in this age group

Nursing-led education and assessment are particularly important for older patients

Copies of this ePoster obtained through Quick Response (QR) codes are for personal use only and may not be reproduced without written permission of the authors.

Plain Language Summary | **ePoster Download** | **Video**

References:
1. Graff JN et al. *Ann Oncol*. 2016;27:286–294.
2. De Giorgi U et al. *Eur J Cancer*. 2021;159:237–246.
3. Armstrong AJ et al. *J Clin Oncol*. 2022;40:1616–1622.

Funding: This study was sponsored by Pfizer and Astellas Pharma, the co-developers of enzalutamide.
Acknowledgments: Medical writing and editorial support funded by the sponsors was provided by Matthieu Larochelle of Onyx, a Prime Global agency.
Note: The data in this abstract were presented at the ASCO Annual Meeting (June 2022) but have not yet been presented with the discussion of and in the context of nursing clinical management.

Contact: nikkidwill@gmail.com
This presentation is intended for a healthcare provider audience.
Presented at the SUNA uroLogic Conference, Hyatt Regency, New Orleans, LA, September 29–October 2, 2022

Background and Methods

- Clinical trials of enzalutamide in different prostate cancer stages have demonstrated a survival benefit in older adults:
 - A subgroup analysis in metastatic castration-resistant prostate cancer (mCRPC) (PREVAIL [NCT01212991]) in men ≥75 years of age found improved median overall survival (OS) with enzalutamide vs placebo (32.4 vs 25.1 months; hazard ratio [HR] 0.61; 95% confidence interval [CI] 0.47, 0.79)[1]
 - In a *post hoc* analysis of PROSPER (NCT02003924), enzalutamide plus ADT was associated with a significant OS benefit irrespective of age group <70 vs ≥70 years in nonmetastatic castration-resistant prostate cancer (nmCRPC) with rapidly rising prostate-specific antigen (PSA)[2]
- In the phase 3 ARCHES trial (NCT02677896), patients with mHSPC received enzalutamide (160 mg/day) plus ADT or placebo plus ADT, and were stratified by disease volume at enrollment and prior docetaxel use – enzalutamide plus ADT provided significant long-term survival benefit vs placebo plus ADT[3]
- A *post hoc* analysis of the ARCHES trial investigated clinical outcomes and safety for two age subgroups: <75 and ≥75 years of age
- Here we review both these data and optimal nursing care (education and clinical management) of patients ≥75 years of age with mHSPC

Results

- In the ARCHES randomized controlled trial (n=1150), 339 patients (29.5%) were ≥75 years of age (enzalutamide plus ADT, n=170; placebo plus ADT, n=169)
 - 133 patients (32.7%) in the <75 years age group and 47 (27.8%) in the ≥75 years age group in the placebo plus ADT arm crossed over to enzalutamide plus ADT after the primary analysis and study unblinding
- The median follow-up time was 44.6 months in the <75 years age group and 44.3 months in the ≥75 years age group
- Clinical outcomes were similar between age groups (**Figure 1**)
- In those ≥75 years of age, enzalutamide plus ADT (compared with placebo plus ADT) demonstrated improvement in time to first symptomatic skeletal event, castration resistance, and PSA progression (**Figure 1B**)
 - After adjusting for possible crossover effects, enzalutamide plus ADT also reduced the risk of death (HR 0.66; 95% CI 0.44, 0.95)
- Due to the *post hoc* nature of the analysis, results should be interpreted with caution

Figure 1. Clinical outcomes in patients with mHSPC, A) <75 and B) ≥75 years of age

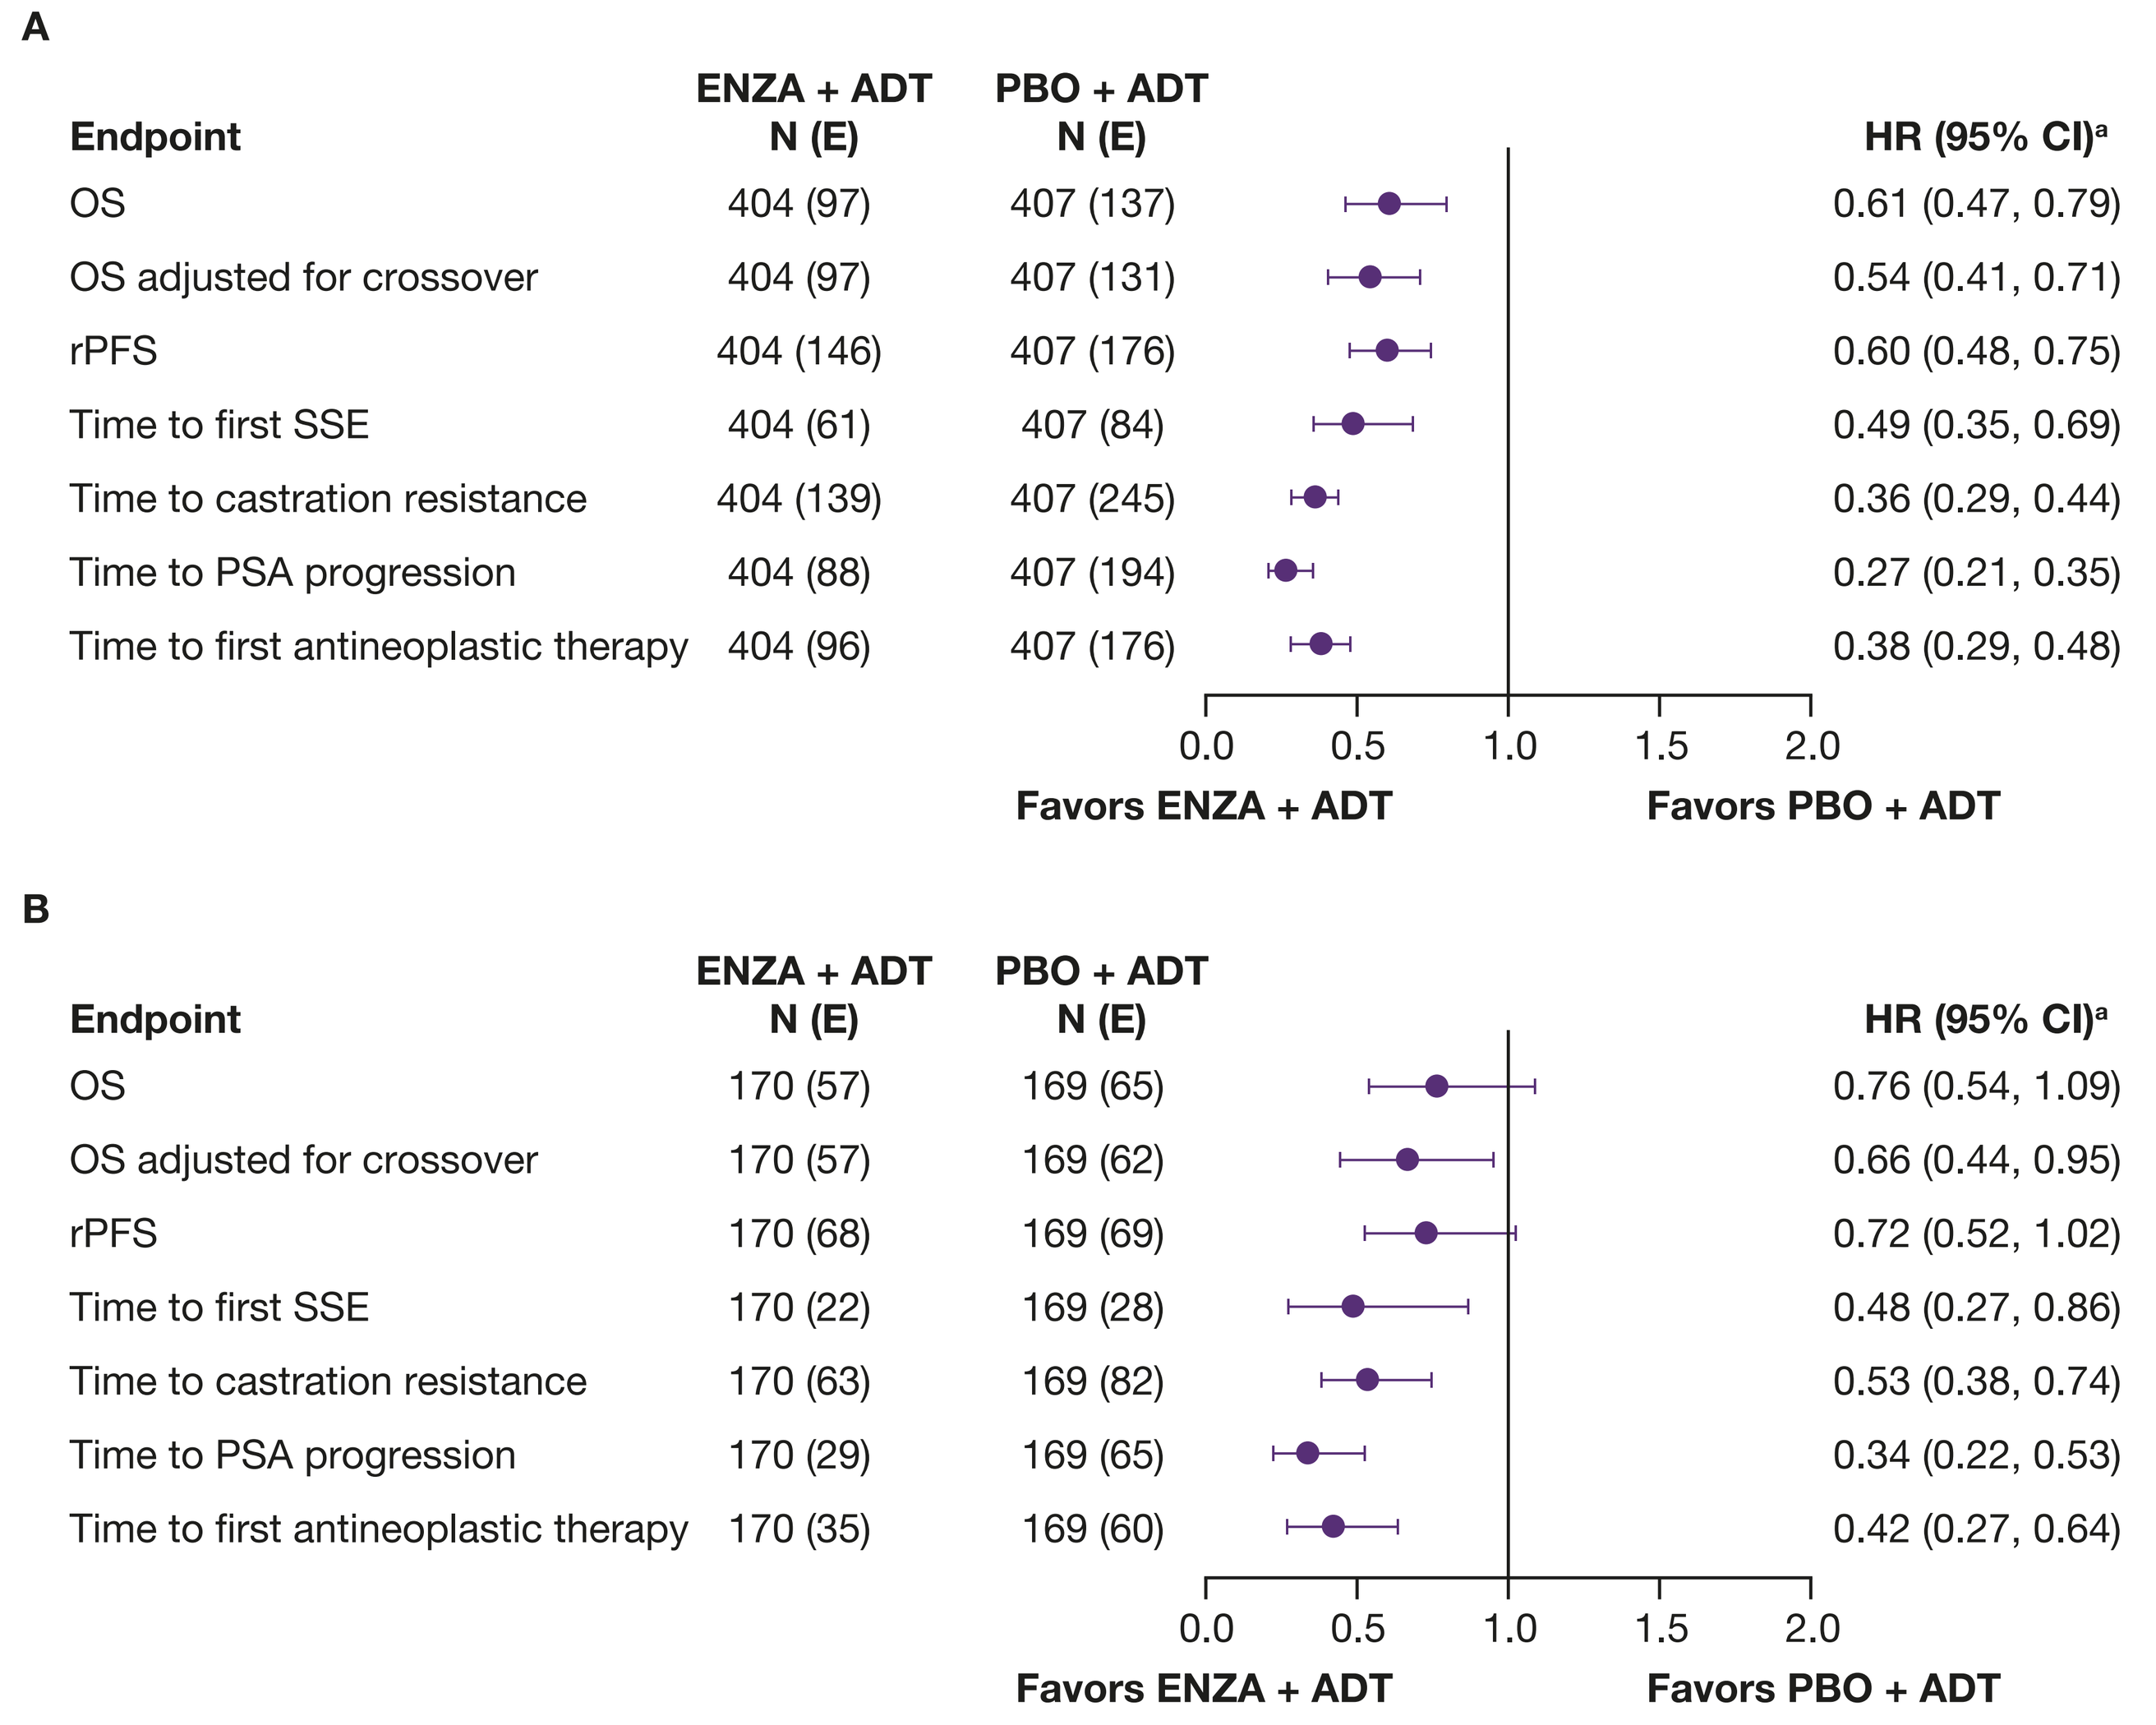
A
Endpoint | ENZA + ADT N (E) | PBO + ADT N (E) | HR (95% CI)[a]
OS | 404 (97) | 407 (137) | 0.61 (0.47, 0.79)
OS adjusted for crossover | 404 (97) | 407 (131) | 0.54 (0.41, 0.71)
rPFS | 404 (146) | 407 (176) | 0.60 (0.48, 0.75)
Time to first SSE | 404 (61) | 407 (84) | 0.49 (0.35, 0.69)
Time to castration resistance | 404 (139) | 407 (245) | 0.36 (0.29, 0.44)
Time to PSA progression | 404 (88) | 407 (194) | 0.27 (0.21, 0.35)
Time to first antineoplastic therapy | 404 (96) | 407 (176) | 0.38 (0.29, 0.48)
0.0 0.5 1.0 1.5 2.0
Favors ENZA + ADT
Favors PBO + ADT
B
Endpoint | ENZA + ADT N (E) | PBO + ADT N (E) | HR (95% CI)[a]
OS | 170 (57) | 169 (65) | 0.76 (0.54, 1.09)
OS adjusted for crossover | 170 (57) | 169 (62) | 0.66 (0.44, 0.95)
rPFS | 170 (68) | 169 (69) | 0.72 (0.52, 1.02)
Time to first SSE | 170 (22) | 169 (28) | 0.48 (0.27, 0.86)
Time to castration resistance | 170 (63) | 169 (82) | 0.53 (0.38, 0.74)
Time to PSA progression | 170 (29) | 169 (65) | 0.34 (0.22, 0.53)
Time to first antineoplastic therapy | 170 (35) | 169 (60) | 0.42 (0.27, 0.64)
0.0 0.5 1.0 1.5 2.0
Favors ENZA + ADT
Favors PBO + ADT

ADT=androgen deprivation therapy; CI=confidence interval; E=number of events; ENZA= enzalutamide; HR=hazard ratio; OS=overall survival; PBO=placebo; PSA=prostate-specific antigen; rPFS=radiographic progresssion-free survival; SSE=symptomatic skeletal event.
[a]HR <1 favors ENZA + ADT; HR >1 favors PBO + ADT. With the exception of the endpoint 'OS adjusted for crossover,' results are not adjusted for crossover, which may potentially impact the results by weakening the benefit of ENZA + ADT treatment. Results are based on the final analysis with a data cut-off date of May 28, 2021.

Treatment Exposure and Safety

- Treatment duration with enzalutamide plus ADT was longer in patients <75 years of age
- Dose reductions and interruptions were more frequent in the older age subgroup in the enzalutamide plus ADT arm (**Table 1**)
- The safety profile of enzalutamide plus ADT and placebo plus ADT was generally similar in the age groups (**Tables 1 and 2**)
- Treatment-emergent adverse events (TEAEs) in the enzalutamide plus ADT arm were more common among those ≥75 years of age; however, the frequency of study-drug-related TEAEs was similar between age groups (**Table 1**)

Table 1. Treatment duration, dose changes, and frequency of TEAEs in patients <75 and ≥75 years of age with mHSPC

	<75 years		≥75 years	
	ENZA + ADT (n=404)	PBO + ADT (n=405)	ENZA + ADT (n=168)	PBO + ADT (n=169)
Median treatment duration, months (range)	41.3 (0.2–58.1)	13.9 (0.4–27.6)	25.9 (0.8–57.8)	13.8 (0.2–27.6)
Dose reduction, n (%)	29 (7.2)	10 (2.5)	20 (11.9)	8 (4.7)
Dose interruption, n (%)	44 (10.9)	29 (7.2)	34 (20.2)	11 (6.5)
Any TEAE, n (%)	360 (89.1)	358 (88.4)	160 (95.2)	146 (86.4)
Any grade 3–4 TEAE, n (%)	146 (36.1)	109 (26.9)	78 (46.4)	51 (30.2)
Any TEAE leading to death, n (%)	14 (3.5)	7 (1.7)	16 (9.5)	5 (3.0)
Any study-drug-related TEAE, n (%)	241 (59.7)	186 (45.9)	98 (58.3)	87 (51.5)
Any study-drug-related TEAE leading to death, n (%)	0	0	0	1 (0.6)

ADT=androgen deprivation therapy; ENZA=enzalutamide; mHSPC=metastatic hormone-sensitive prostate cancer; PBO=placebo; TEAE=treatment-emergent adverse event.

- In the ≥75 years of age group, some TEAEs were more common among patients receiving enzalutamide plus ADT vs placebo plus ADT (**Table 2**)
- However, these events occurred at higher rates in the older age subgroup in both the treatment arms, and with the exception of loss of consciousness, relative rates between age subgroups for enzalutamide vs placebo were similar

Table 2. Exposure-adjusted TEAEs of special interest in patients <75 and ≥75 years of age with mHSPC[a]

	<75 years				≥75 years			
	ENZA + ADT (n=404)		PBO + ADT (n=405)		ENZA + ADT (n=168)		PBO + ADT (n=169)	
	n (%)	Rate[b]	n (%)	Rate[b]	n (%)	Rate[b]	n (%)	Rate[b]
Musculoskeletal events	164 (40.6)	14.60	129 (31.9)	24.75	59 (35.1)	14.83	41 (24.3)	19.33
Fracture	53 (13.1)	4.72	20 (4.9)	3.84	24 (14.3)	6.03	11 (6.5)	5.19
Fall	29 (7.2)	2.58	9 (2.2)	1.73	29 (17.3)	7.29	10 (5.9)	4.72
Fatigue	129 (31.9)	11.48	82 (20.2)	15.73	55 (32.7)	13.83	36 (21.3)	16.97
Cognitive/memory impairment	24 (5.9)	2.14	9 (2.2)	1.73	14 (8.3)	3.52	6 (3.6)	2.83
Hepatic disorder	26 (6.4)	2.31	28 (6.9)	5.37	8 (4.8)	2.01	6 (3.6)	2.83
Hypertension	66 (16.3)	5.87	27 (6.7)	5.18	16 (9.5)	4.02	12 (7.1)	5.66
Ischemic heart disease	17 (4.2)	1.51	5 (1.2)	0.96	9 (5.4)	2.26	6 (3.6)	2.83
Other selected CV events	15 (3.7)	1.33	3 (0.7)	0.58	10 (6.0)	2.51	7 (4.1)	3.30
Loss of consciousness	5 (1.2)	0.44	1 (0.2)	0.19	10 (6.0)	2.51	1 (0.6)	0.47

ADT=androgen deprivation therapy; CV=cardiovascular; ENZA=enzalutamide; mHSPC=metastatic hormone-sensitive prostate cancer; PBO=placebo; TEAE=treatment-emergent adverse event.
[a]Those occurring in ≥5% in either treatment arm in at least one of the age subgroups.
[b]Calculated as (number of events/total exposure in patient-years) x 100.
Note: The following occurred in <5% in both age subgroups in each treatment arm and at similar rates in the enzalutamide and placebo arms: rash, second primary malignancies, renal disorder, neutrophil count decreased, angioedema, thrombocytopenia, convulsions, severe cutaneous adverse reactions, and posterior reversible encephalopathy syndrome.

Implications for Nursing Practice

Monitoring and Assessment

- Patients with mHSPC receiving enzalutamide plus ADT should be monitored for signs and symptoms of AEs and disease progression
 - Patients ≥75 years of age may be at increased risk for falls and cognitive impairment
 - Muscle weakness or stiffness, uneven gait, and vision, hearing, or balance impairment can be signs of fall risk
 - Ask about and assess for cognitive decline to ensure accurate home medication administration and adherence
 - Note when refills are requested (earlier or later than expected), which may indicate missed doses or overdosing
- Home medications should be reconciled at each visit (including dose changes); over-the-counter medications, herbals, supplements, and vitamins should be reported as well
- Assess any signs, symptoms, and abnormal laboratory results during both in-person and virtual interactions and communicate findings to the multidisciplinary care team
- Monitor labs (including PSA) and bone density, and ensure adequate dietary or supplementary calcium and vitamin D
- When caring for patients ≥75 years of age who have pre-existing cardiovascular disease, vital signs and blood pressure should be monitored and the cardiology or primary care teams should be engaged to optimally manage the patient's cardiovascular disease
- Gastrointestinal issues in older adults can impact medication adherence; appetite and weight should be monitored

Patient Education and Support

- Nurses are a valuable source of support and information about enzalutamide treatment for patients and their caregivers
- Patients should be advised to take enzalutamide at the same time daily, and, in the case of a missed dose, the next dose should *not* be increased
- It is important that nurses educate patients on the mechanism of action, AEs, and potential medication interactions of enzalutamide
- Patients and caregivers should be advised on fall prevention measures, proper exercise, and nutrition
 - The patient and family should be advised to clear the home of clutter, and the patient should be advised to wear eyeglasses and hearing aids and use assistive devices as needed, exercise to maintain strength, and stay well-hydrated if they have been determined to have a high fall risk

Conclusion

- These data show that enzalutamide plus ADT is efficacious and safe in the older mHSPC population of ARCHES, similar to what was observed in CRPC from the PROSPER and PREVAIL trials
- However, elderly patients may have some increased side effects, which may require additional safety monitoring
- Patient education may be particularly important for patients age 75 and older

Copyright ©2022. All rights reserved.